Optimisation of Sample Preparation in the Particle-Size Characterization of Active Pharmaceutical Ingredients by Laser Diffraction

Maria Ellul [1], Oliver Schembri [2] and Claude Farrugia [1]

[1]Department of Chemistry, University of Malta, Msida MSD2080, Malta

[2]Arrow Pharm (Malta) Ltd., HF 62, Ħal Far Industrial Estate Birżebbuġa BBĠ 3000

Introduction

The particle size of active pharmaceutical ingredients (API's) can affect medicinal product processability, manufacturability, and biopharmaceutical characteristics. Thus, it is imperative to monitor mean particle size and particle size distribution to establish particle size supplier specifications. Laser diffraction is currently the dominant method of particle size distribution analysis, since it is a fast and non-destructive technique, which may be applied to an array of particulate systems presented in various physical forms. However, successful particle size measurement depends on the development of appropriate sampling, sample preparation and measurement techniques. Spatulation, a dispersion technique wherein small portions of the API powder are mixed with small portions of the dispersant, and worked in together using a spatula until all of the components are uniformly combined and blended, suffers from operator variability. The objective of this study was therefore to optimise the method of preparation of two API dispersions, by eliminating the need for spatulation.

Methodology

The two API's used in this study were *ramipril* and *terbinafine hydrochloride.* Laser diffraction measurements were performed with a Malvern Mastersizer 2000. A number of dispersant-surfactant combinations were selected and their inertness at inducing changes to the API particles by dissolution was monitored using IR spectroscopy. The dispersant-surfactant combination that gave the slowest rate of settling of the dispersed particles for each API was established by monitoring the absorbance of the samples as a function of time at 600 nm in a UV-Vis spectrophotometer. The particles which stayed in suspension for the longest time period, as established from the stability of the turbidimetric readings, were taken to represent a well-dispersed suspension. Furthermore, sonication of the sample was employed to ensure that any agglomerates or large clumps were fully dispersed to the primary particle size of the material. Sonication time was varied until the particle size, as monitored by laser diffraction, did not change with further increase in sonication time. Sample-to-sample variation (repeatability) was examined across six samples, and two other batches of the API in question, ensuring reproducibility of the technique.

Results

Figure 1: *Ramipril* - *Top Graph*: Within batch variability before method optimization. *Bottom Graph*: Within batch variability after method optimization.

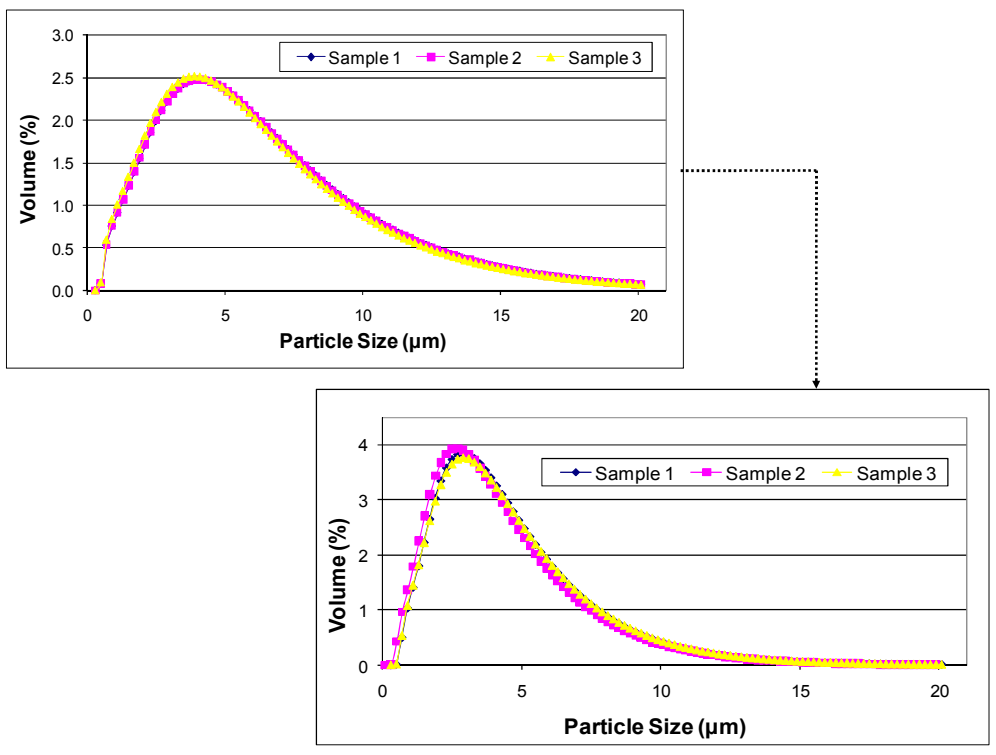
Sample 1
Sample 2
Sample 3
Volume (%)
Particle Size (µm)

Figure 2: *Terbinafine HCl – Top Graph*: Within batch variability before method optimization. *Bottom Graph*: Within batch variability after method optimization.

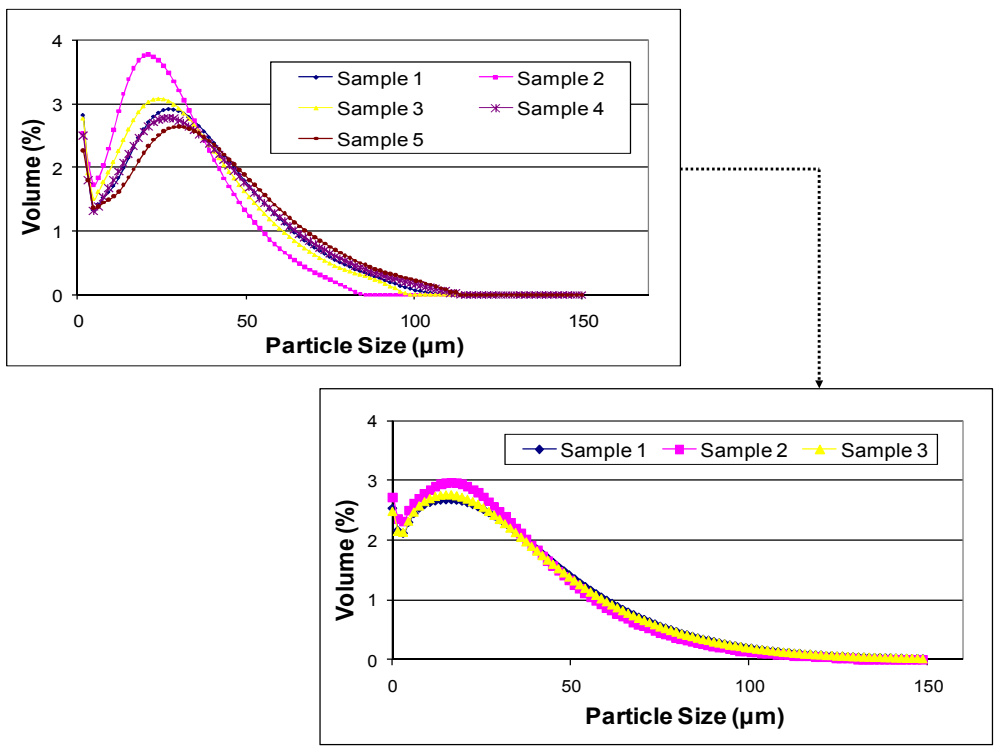
Sample 1
Sample 2
Sample 3
Sample 4
Sample 5
Volume (%)
Particle Size (µm)

Ramipril was found to disperse best in Isopar G and lecithin. Terbinafine HCl dispersed best in mineral oil and lecithin.

<u>Terbinafine HCl</u>: Visual assessment of dispersion turbidity showed the ideal lecithin concentration to be 0.5% w/v.
<u>Ramipril</u>: Since there was no clear gradation of turbidity, turbidimetry at 600nm was used. The ideal lecithin concentration was found to be 1.0% w/v.

Figure 3: The effect of different surfactant concentrations on ramipril dispersion.

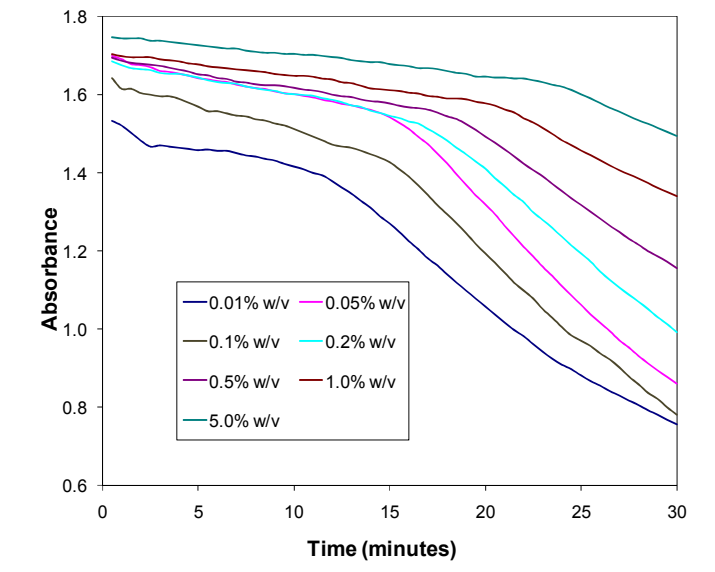
Absorbance
Time (minutes)
0.01% w/v
0.05% w/v
0.1% w/v
0.2% w/v
0.5% w/v
1.0% w/v
5.0% w/v

Figure 4: Dispersion stability at optimal sonication time for *ramipril* (*left*) and *terbinafine HCl* (*right*). A sonication time of 2 minutes was chosen for the *ramipril* dispersion, and 5 minutes was selected for the *terbinafine HCl* dispersion.

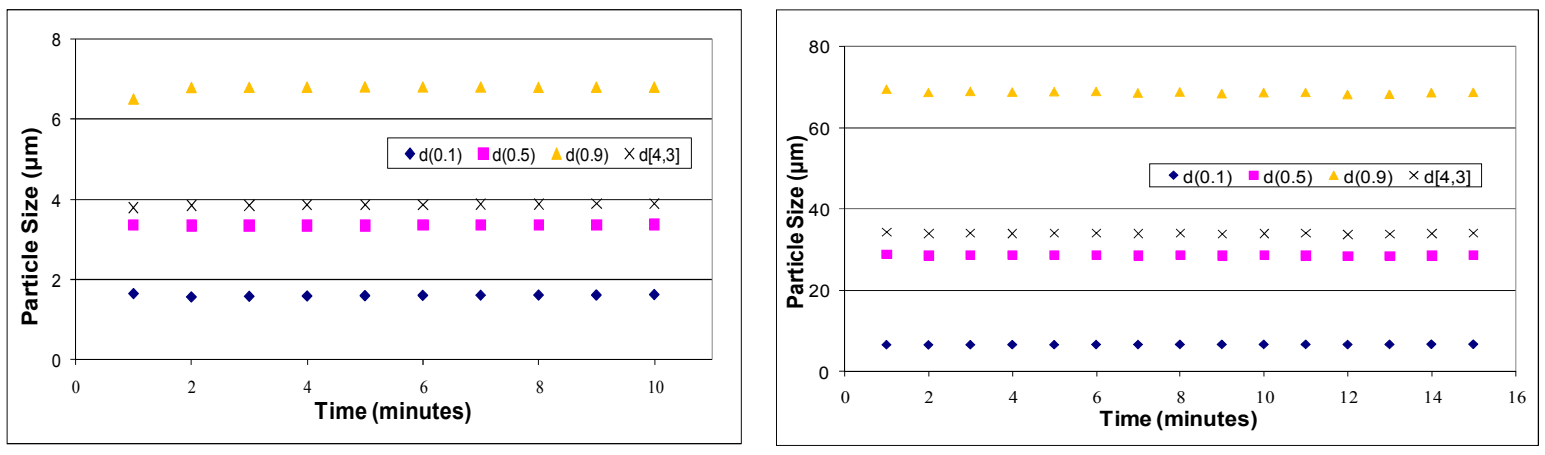
d(0.1)
d(0.5)
d(0.9)
d[4,3]
Particle Size (µm)
Time (minutes)

Conclusions

Although laser diffraction instruments with good specification and verified performance can be employed to accurately characterize many different materials, successful particle size measurement depends on the development of an appropriate method (Rawle and Kippax, 2010).
The method optimization process resulted in a significantly reduced variability in the measured particle size. For *ramipril*, the relative standard deviation (RSD) of the d(0.1) statistic was reduced from 3.148% to 0.557%, and that of the d(0.9) statistic from 1.315% to 1.026%. For *terbinafine HCl*, the value away from the central distribution d(0.1) was reduced from 7.3% to 4.364%, and the value of d(0.9) decreased from 9.8% to 9.446%.
The lack of polarity of the two API's, characterised by the low ratios of polar surface area to molecular surface area *(ramipril*: 95.94 Å^2 / 654.30 Å^2 ; *terbinafine HCl*: 3.24 Å^2 / 493.72 Å^2, as computed by Calculator Plugins [Marvin 5.5.0.0, 2011, ChemAxon, http://www.chemaxon.com]), was considered a key factor in determining the suitability of non-polar dispersing agents and the relatively non-polar surfactant lecithin.

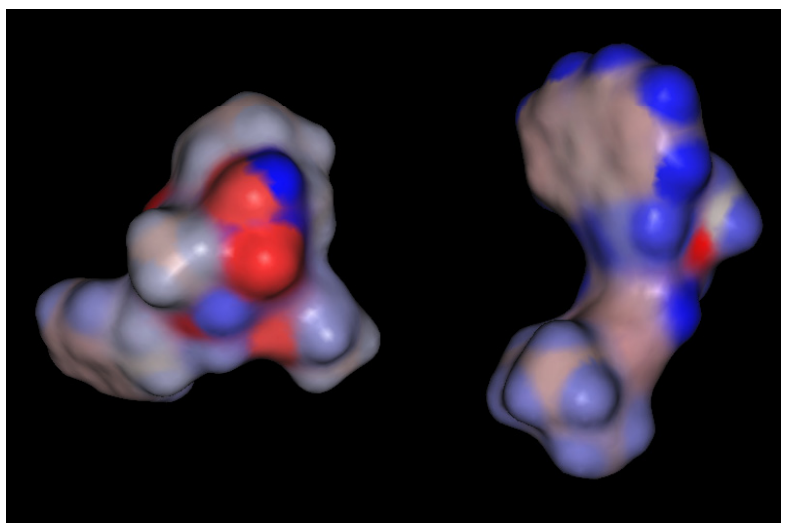

Figure 5: Surface properties of *ramipril* (*left*) and *terbinafine HCl* (*right*), as generated by ChemAxon, showing nonpolar (*white*) and polar (*blue*: positively charged, *red*: negative charged) areas.

References

Rawle, A.; Kippax, P.; *Setting New Standards for Laser Diffraction Particle Size Analysis*. Technical Article MRK1399-01 [Online] **2010**. Malvern Instruments Inc.

Acknowledgements

The authors thank Arrow Pharm Ltd. for the supply of APIs and other chemicals, and for the access to scientific equipment in their laboratories.